Combination of oncolytic vaccinia virus and immune checkpoint blockade overcomes resistance to immunotherapy in renal cell carcinoma

Hong Jae Chon[1,2], Won Suk Lee[1,2], So Jung Kong[1,2], Hannah Yang[1,2], Na Keum Lee[1,2], Intae Park[3], Eun Sang Moon[4], Ji Won Choi[4], Joong Bae Ahn[5], Joo Hang Kim[1], Gou Young Koh[3], Chan Kim[1,2]

[1]Medical Oncology and [2]Laboratory of Tumor Vasculature and Microenvironment, CHA Bundang Medical Center, CHA University, Seongnam, Korea

[3]Graduate School of Medical Science and Engineering, KAIST, Daejeon, Korea, [4]SillaJen, Inc., Seoul, Korea, [5]Yonsei Graduate School, Yonsei University College of Medicine, Seoul, Korea

Introduction

- Recently, immune checkpoint inhibitor (ICI) has emerged as another standard of care in advanced cancer, but response is limited as a monotherapy. To determine the optimal combination strategy for cancer immunotherapy, we employed mJX-594 (hereafter referred to as JX), an oncolytic vaccinia virus, as a combination partner for ICI.
- Here, we comprehensively dissected the changes of tumor microenvironment with mJX-594 virotherapy and investigated its immunotherapeutic potential to provide a rational combinatorial strategy with ICIs in poorly immunogenic tumor models with emphasis on kidney cancer.

Materials and Methods

- **Generation of virus**: mJX-594 was provided by Sillajen, Inc. (Seoul, Korea). Briefly, mJX-594 is a Western Reserve strain of oncolytic vaccinia virus with murine GM-CSF in the vaccinia thymidine kinase gene locus under the control of the p7.5 promoter.
- **Tumor model and treatment regimens**: Tumors were implanted by SC injection of 2 x 10^5 Renca cells into the right flank of mice. When tumor size reached >50 mm^3, mice were treated with 1 x 10^7 pfu of mJX-594 every 3 days. For depletion study, depletion antibodies for $CD4^+$ (200 ug), $CD8^+$ (200 ug) T cells or GM-CSF^+ (200 ug) were injected IP with mJX-594. For immune checkpoint blockade, αPD-1 (10 mg/kg, BioXCell) and/or αCTLA-4 (4 mg/kg, BioXCell) antibodies injected IP every 3 days.

Results

Figure 1. mJX-594 virotherapy converts immunosuppressive non-inflamed tumors into inflamed tumors.

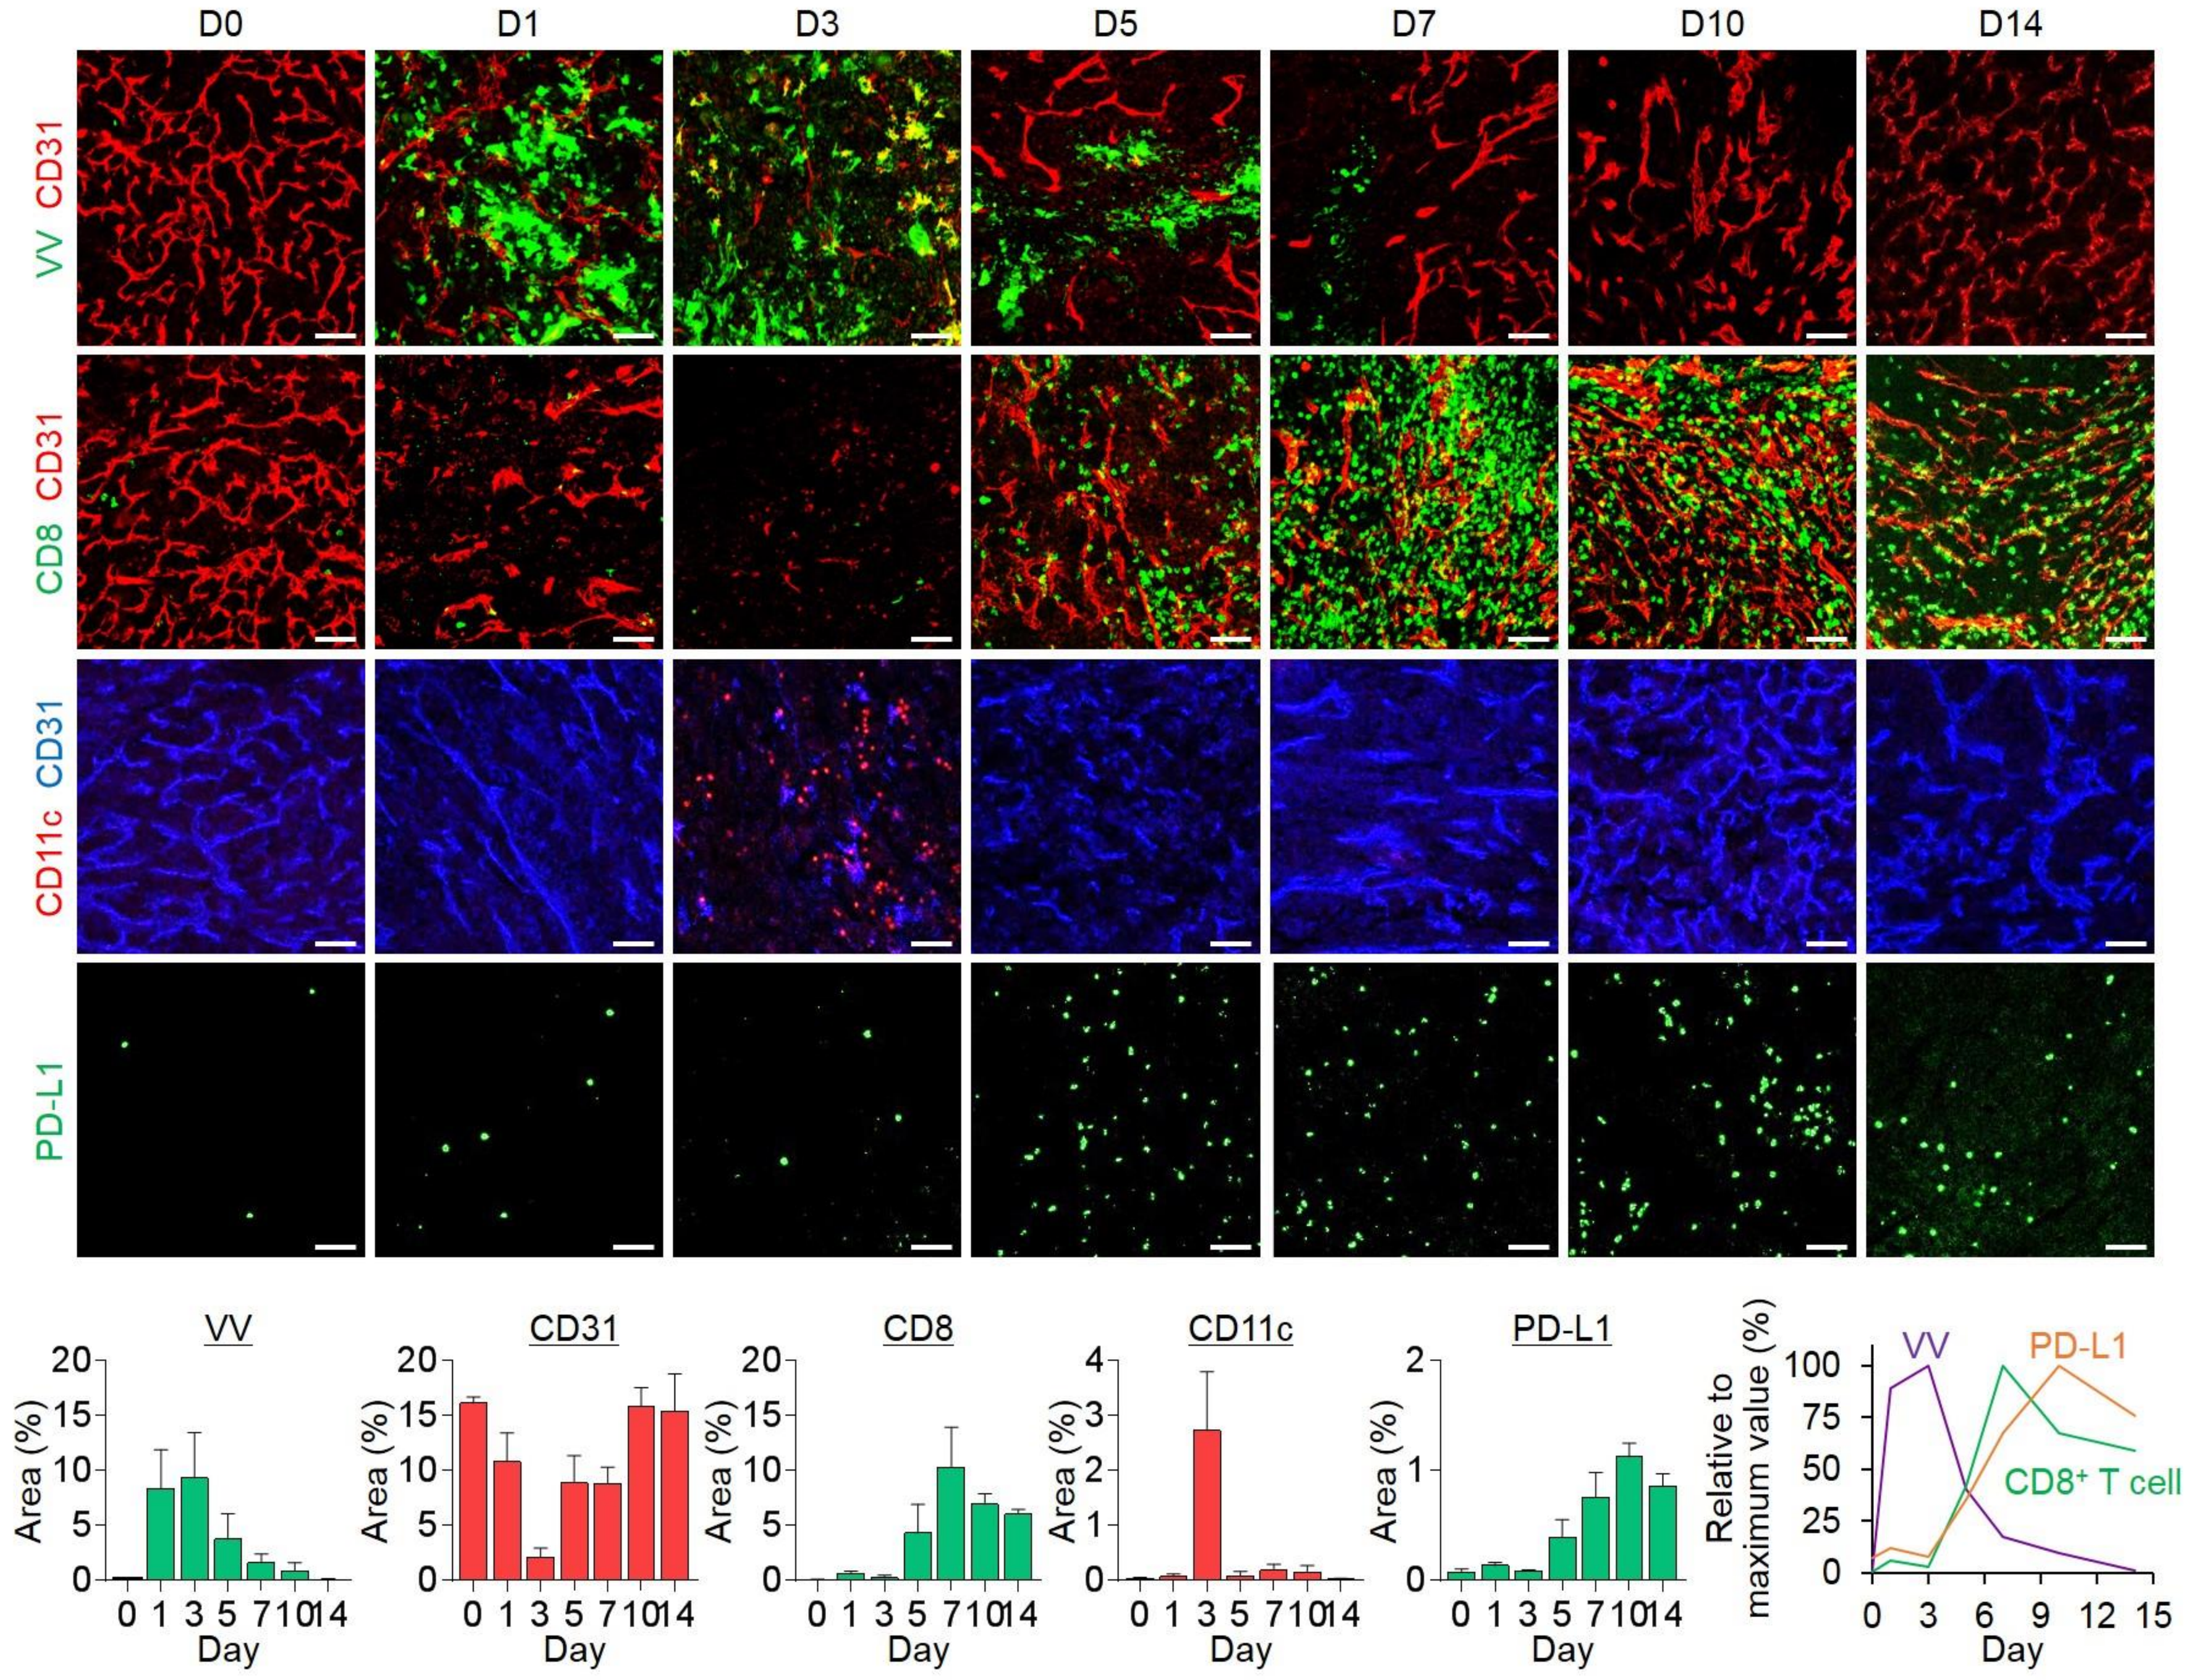
D0
D1
D3
D5
D7
D10
D14
VV CD31
CD8 CD31
CD11c CD31
PD-L1
VV
CD31
CD8
CD11c
PD-L1
Area (%)
Day
Relative to maximum value (%)
PD-L1
CD8+ T cell

VV, Vaccinia virus

Results

Figure 2. PD-L1 is predominantly expressed in tumor cells after mJX-594 virotherapy.

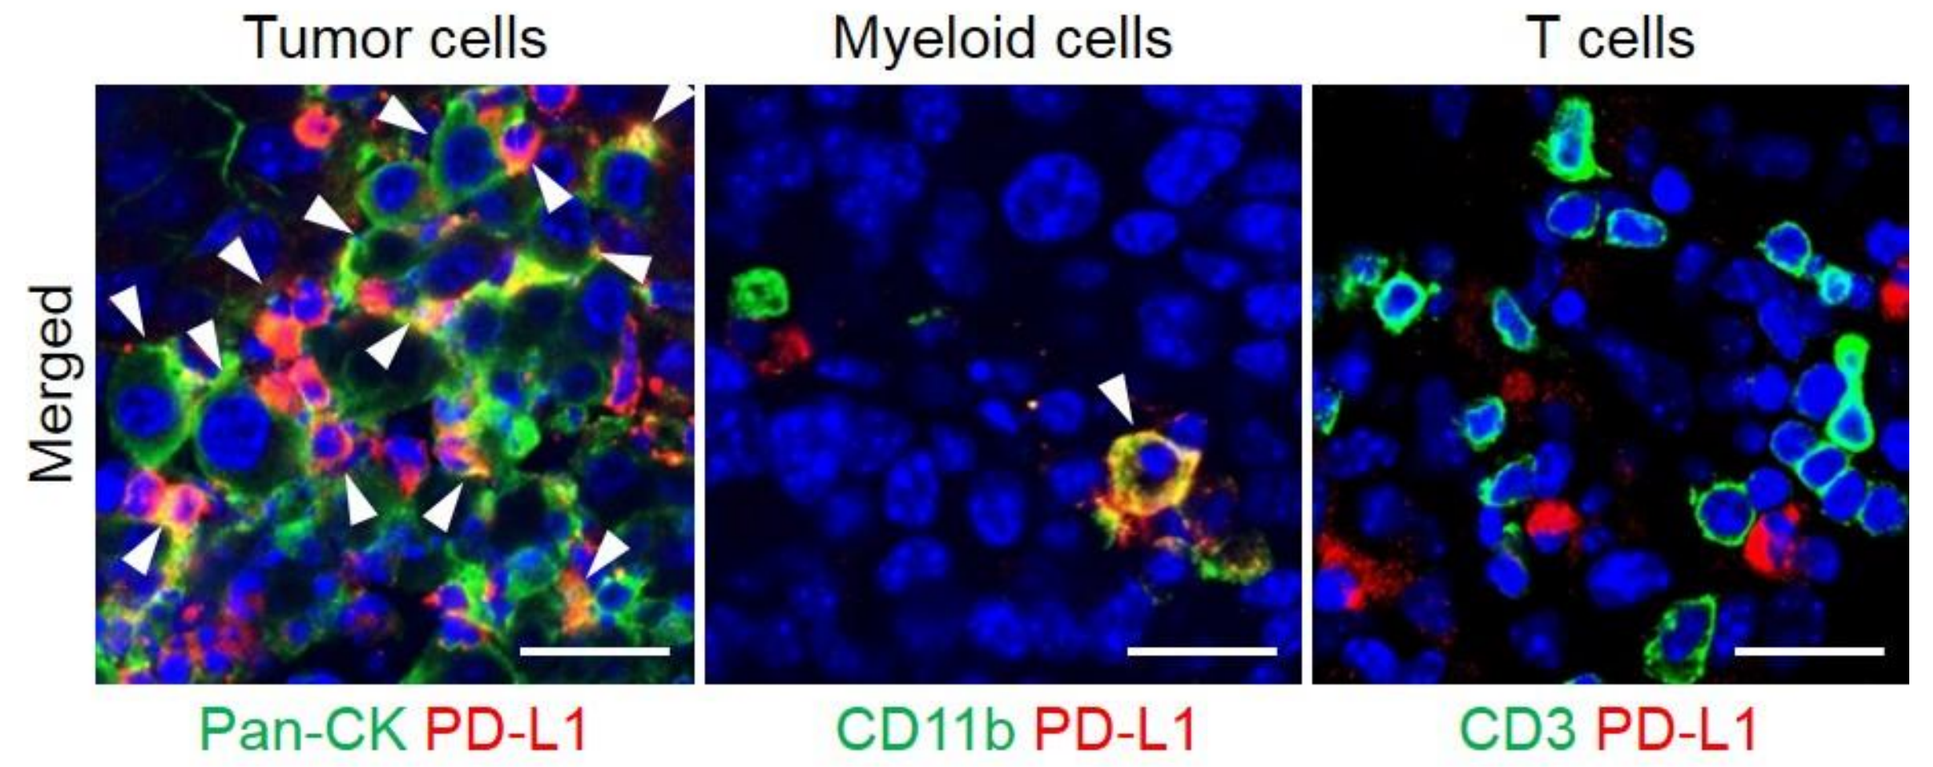
Tumor cells
Myeloid cells
T cells
Merged
Pan-CK PD-L1
CD11b PD-L1
CD3 PD-L1

Figure 3. PanCancer Immune Profiling panel shows remarkable differences in immune signatures between mJX-594-treated tumors and control tumors.

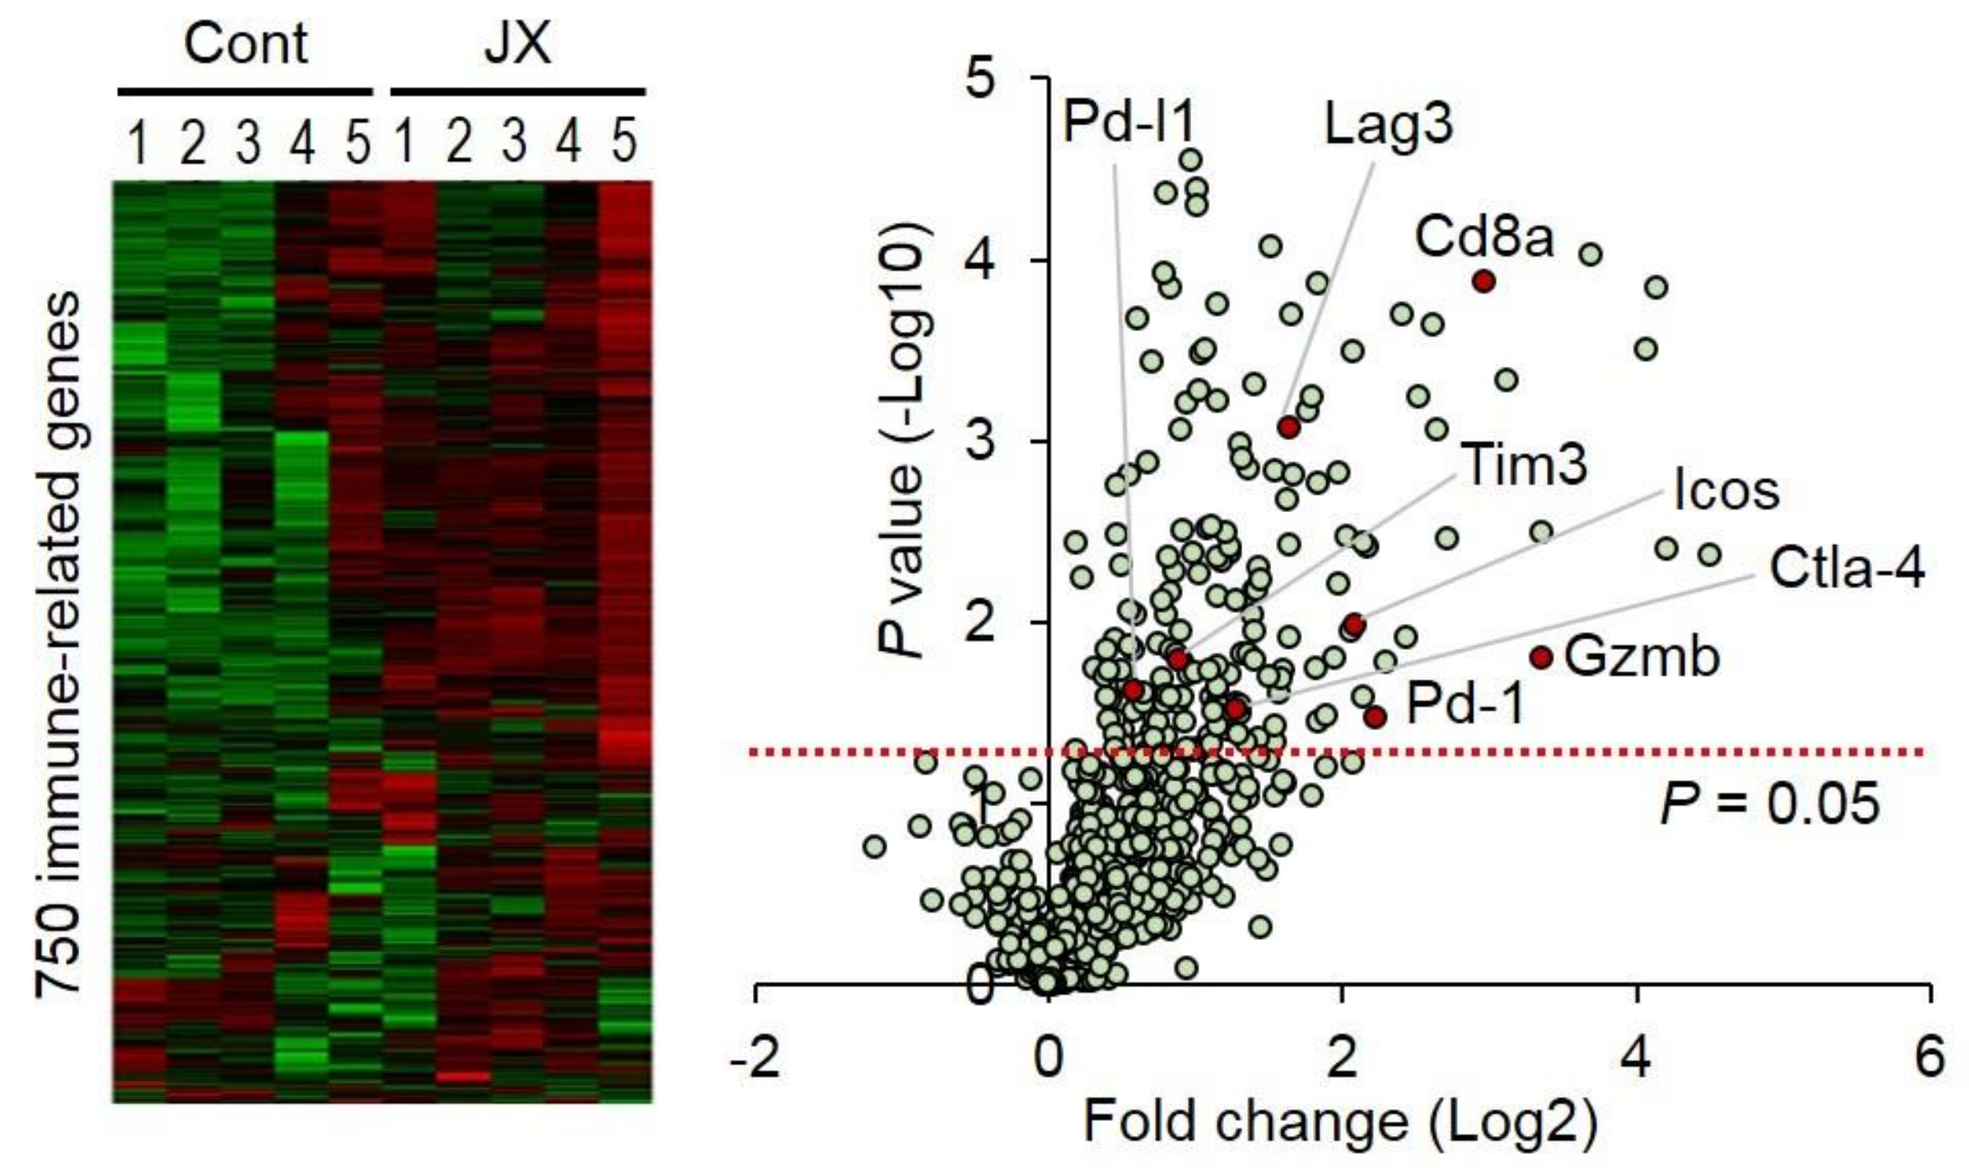
Cont
JX
750 immune-related genes
Pd-l1
Lag3
Cd8a
Tim3
Icos
Ctla-4
Gzmb
Pd-1
P = 0.05
P value (-Log10)
Fold change (Log2)

Figure 4. Intratumoral injection of mJX-594 leads to systemic and cancer-specific immune responses.

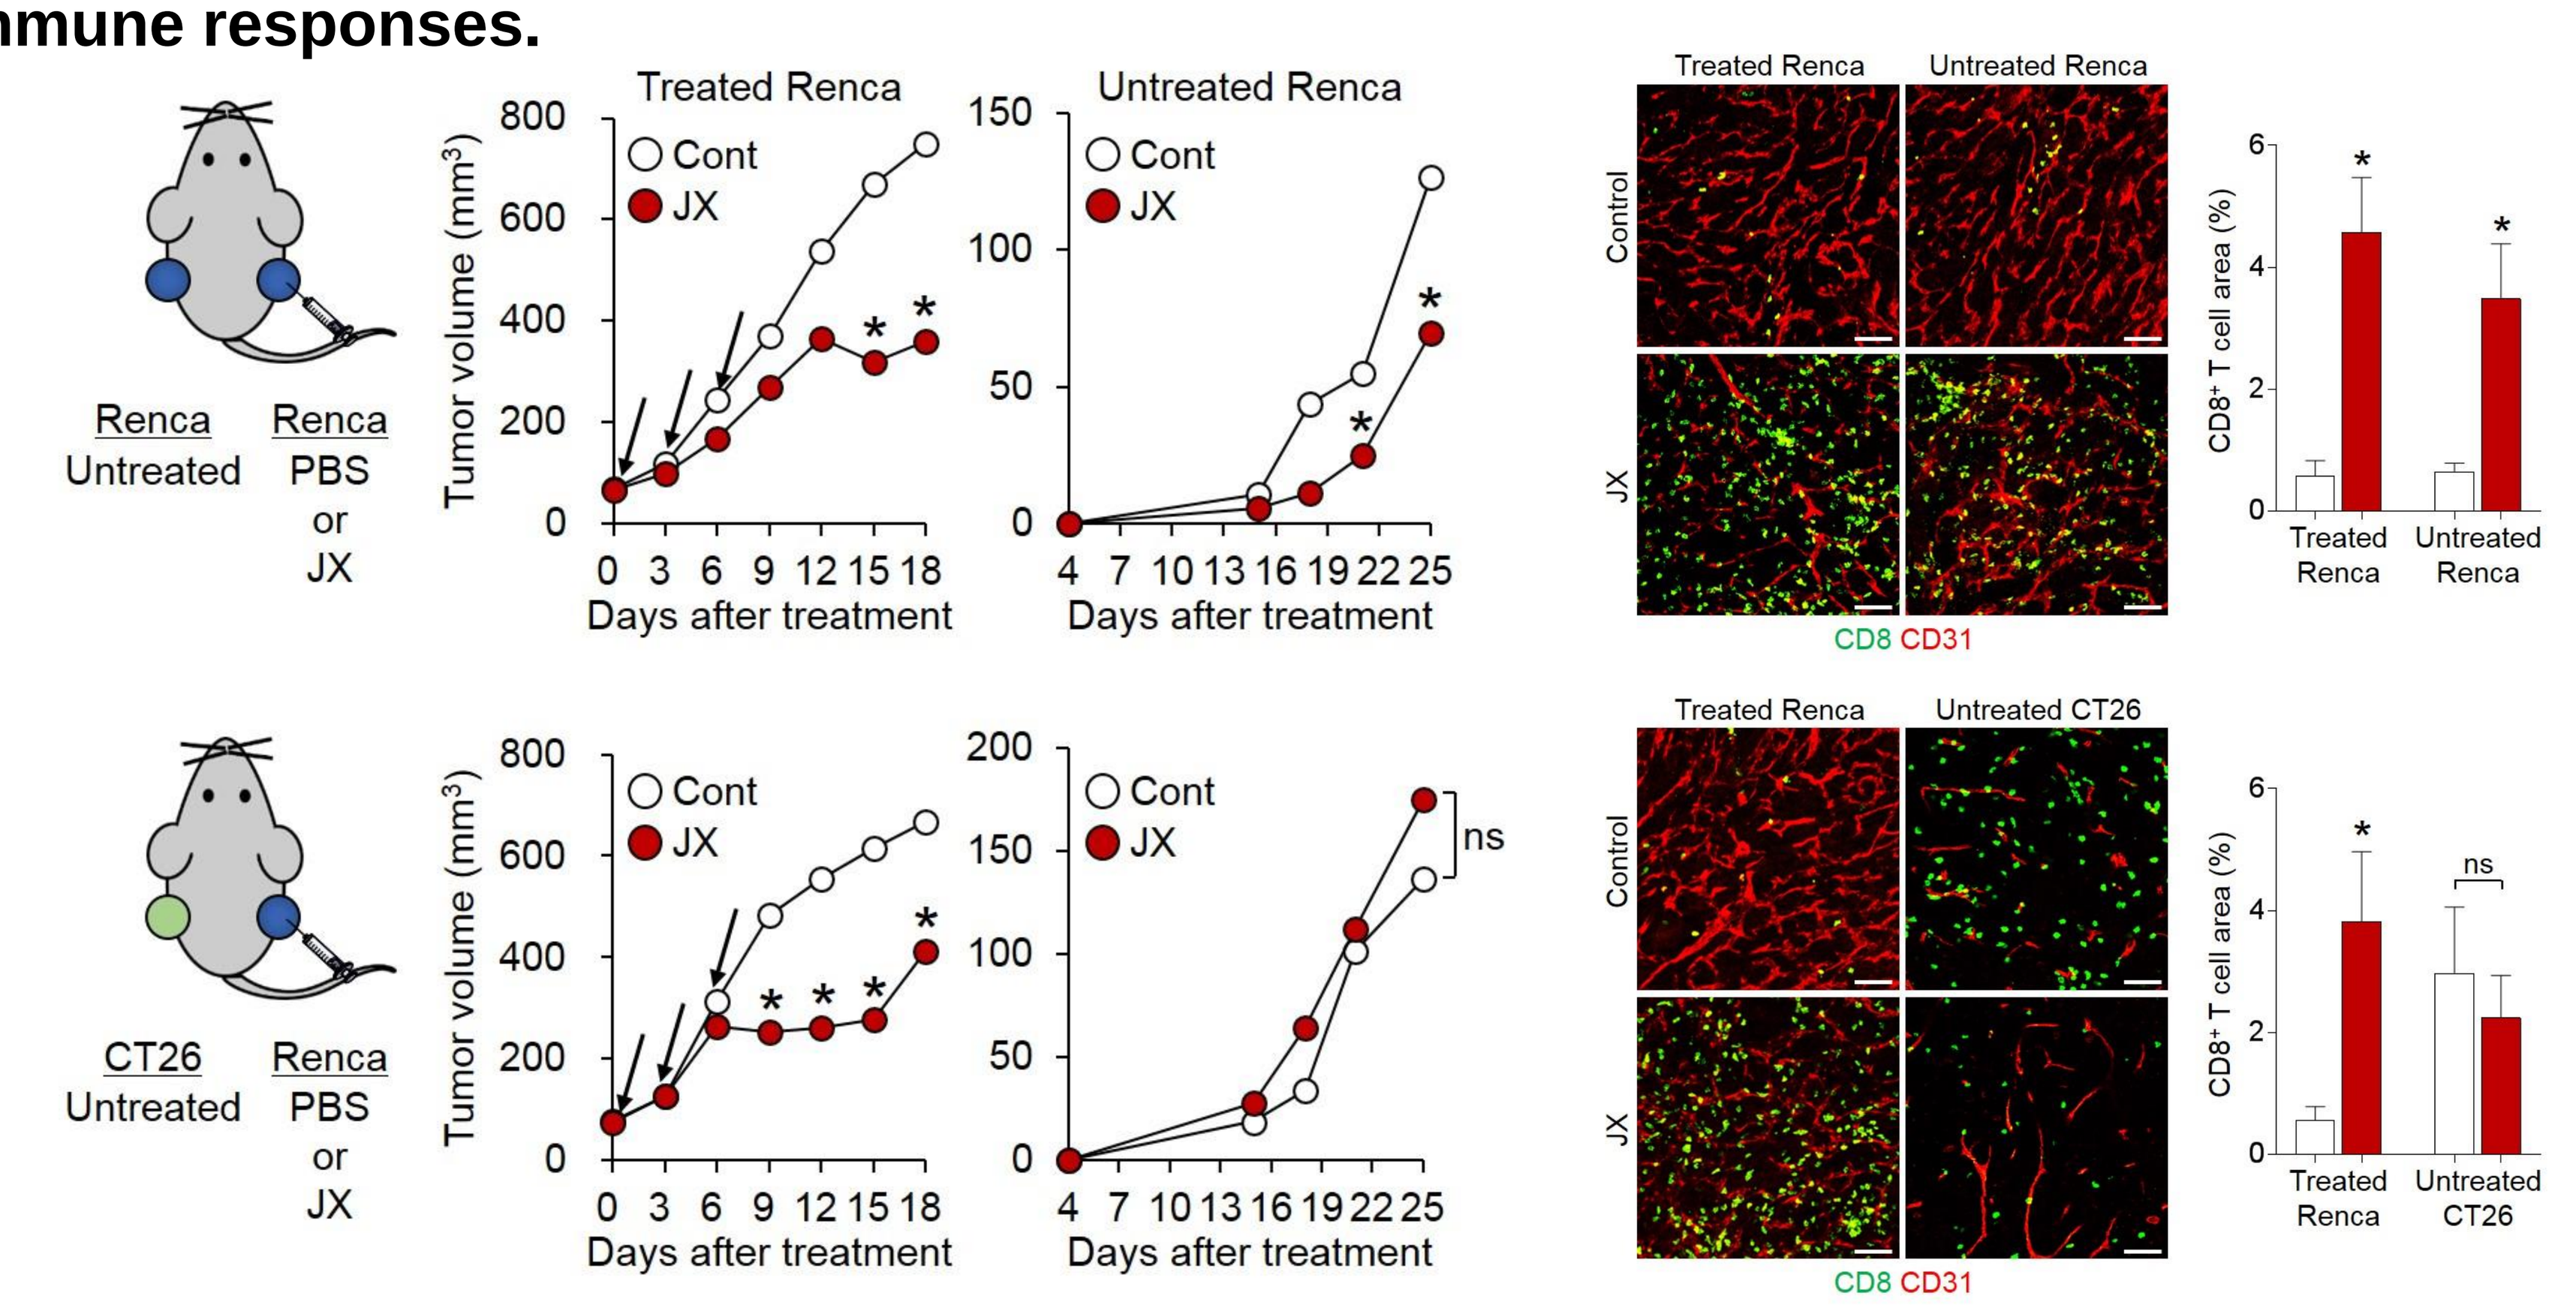
Renca Untreated
Renca PBS or JX
CT26 Untreated
Treated Renca
Untreated Renca
Untreated CT26
Cont
JX
Tumor volume (mm3)
Days after treatment
Control
CD8 CD31
CD8+ T cell area (%)

Figure 5. Depletion of T cells or GM-CSF significantly negated the anti-cancer effect of mJX-594 virotherapy.

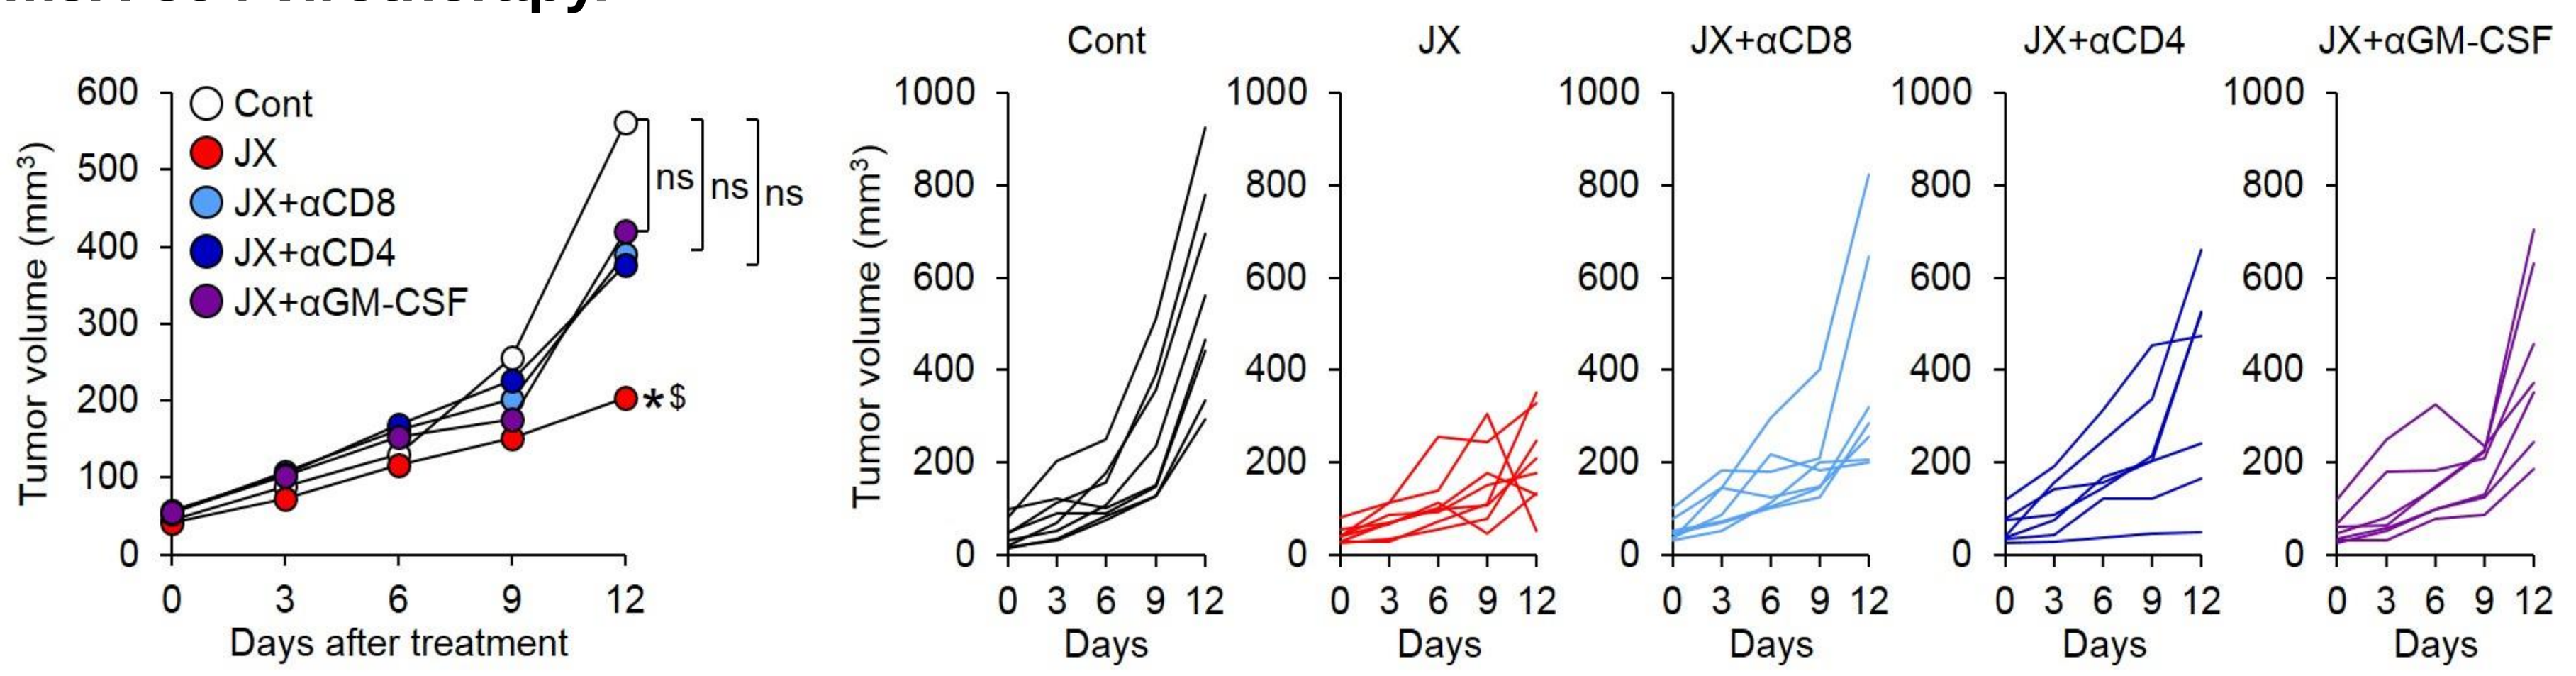
Cont
JX
JX+αCD8
JX+αCD4
JX+αGM-CSF
Tumor volume (mm3)
Days after treatment
Days

*p < 0.05 versus control; $p < 0.05 versus αGM-CSF. ns, not significant.

Figure 6. Combination of mJX-594 with immune checkpoint blockade elicits a synergistic anti-cancer effect with enhanced infiltration of T lymphocytes into tumor.

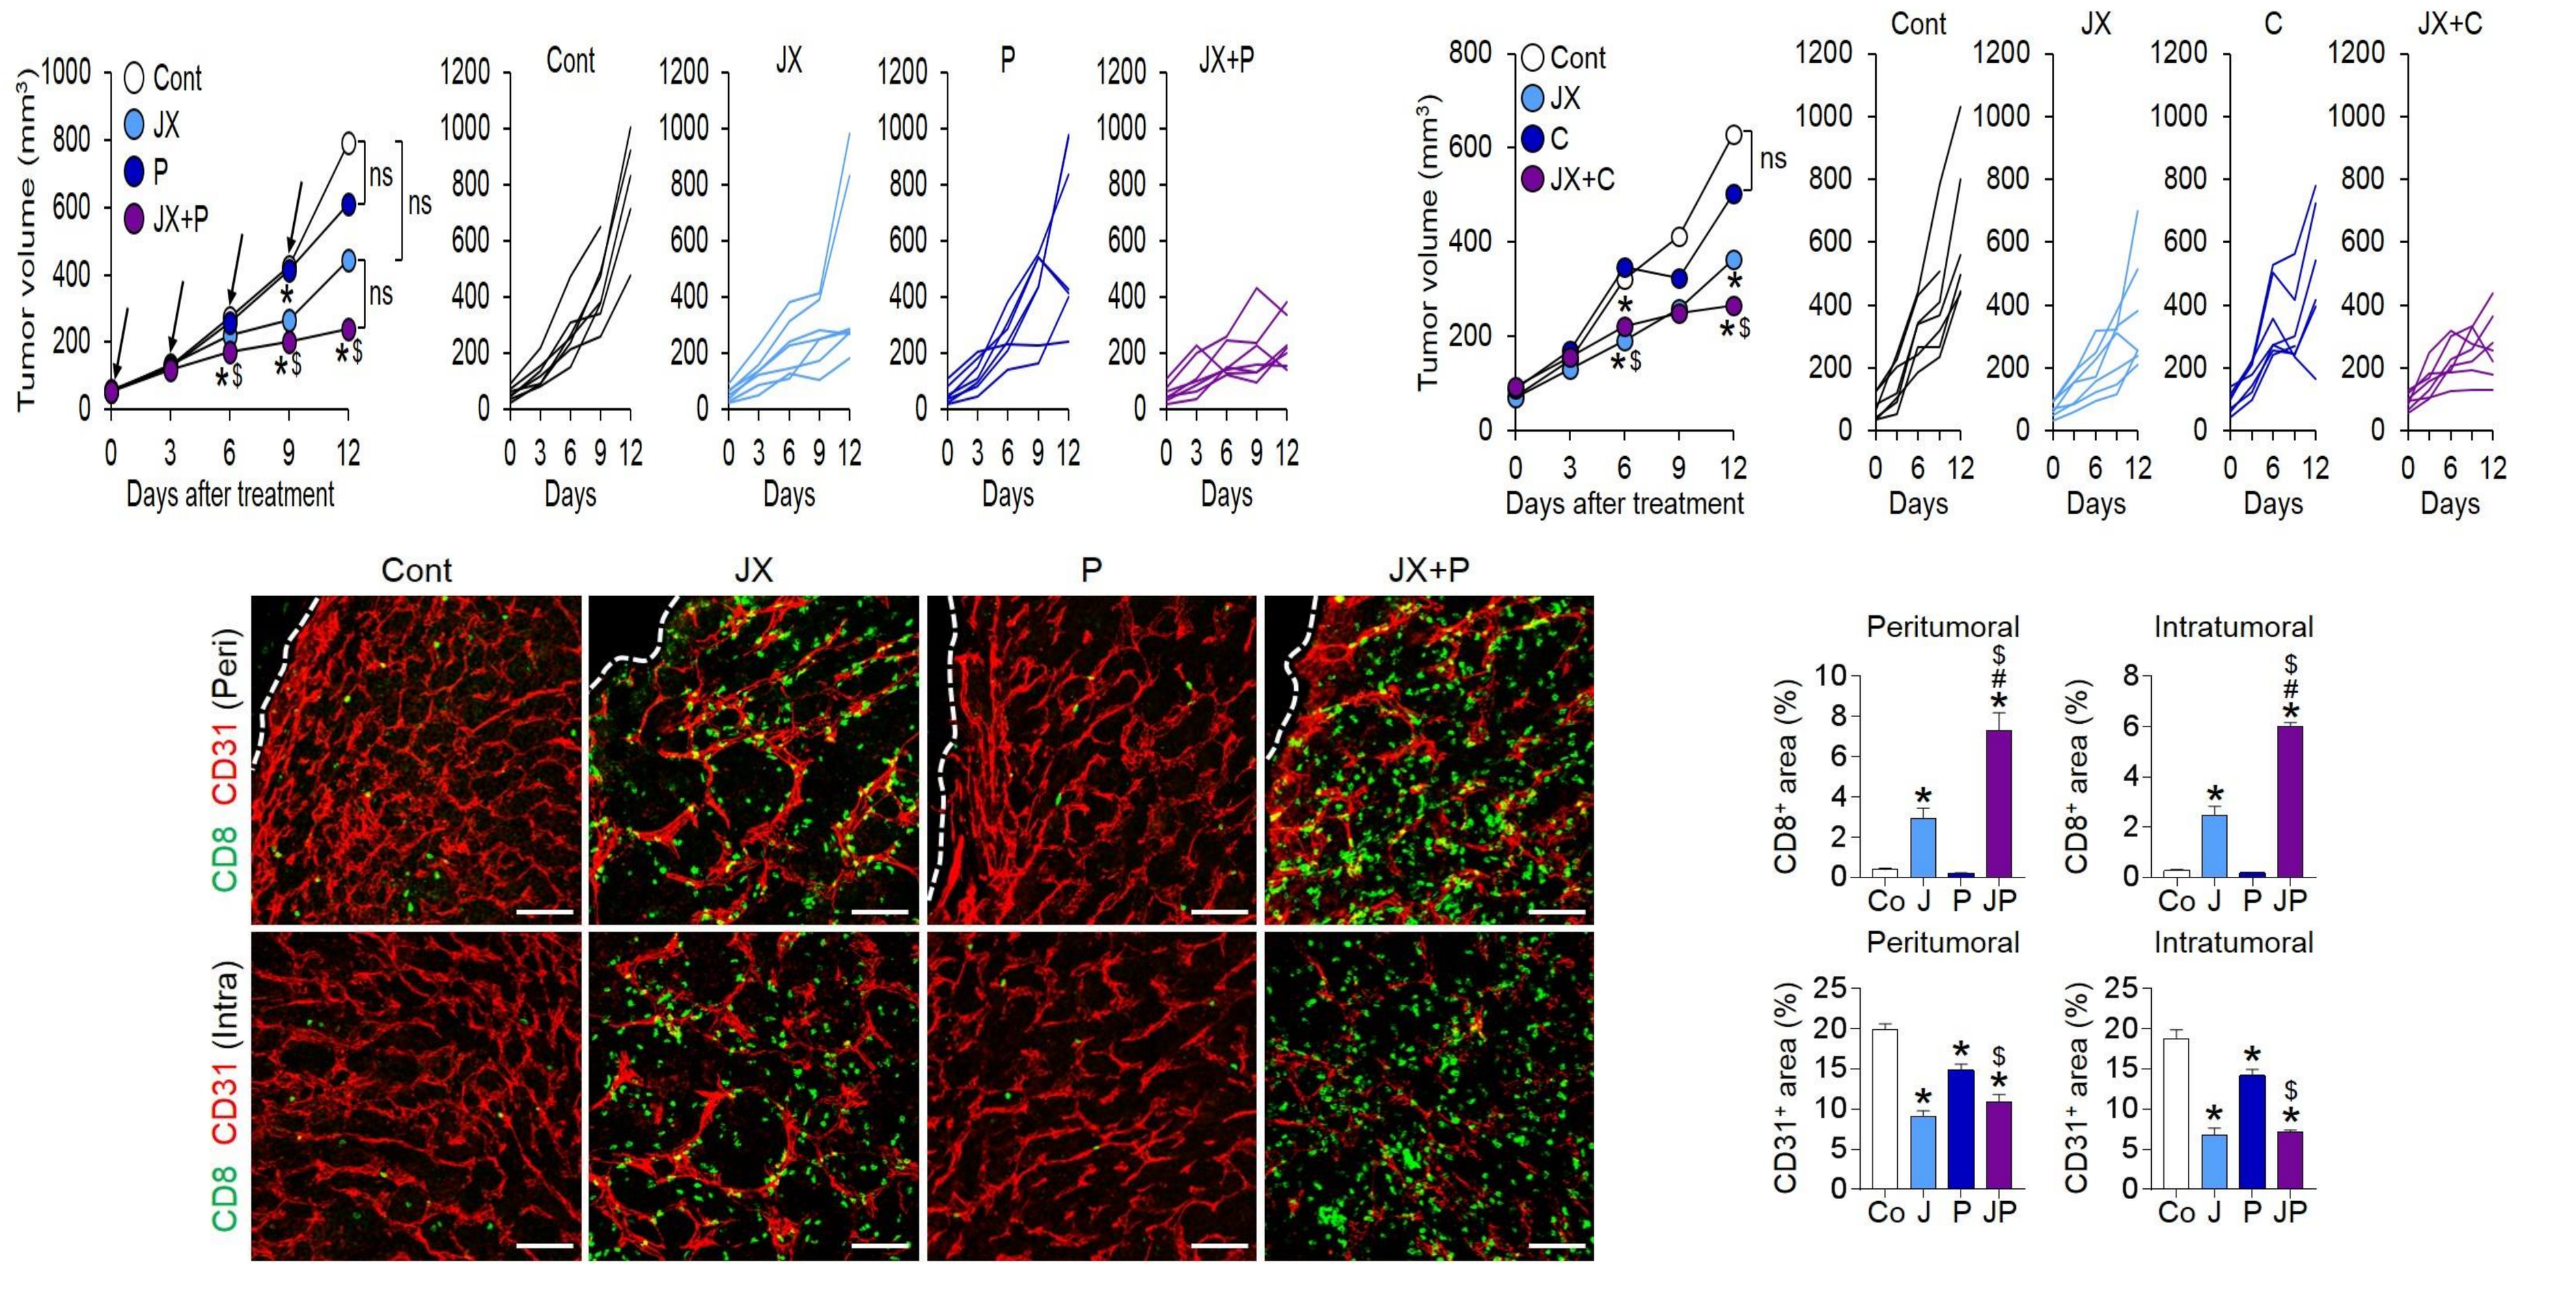
Cont
JX
P
JX+P
C
JX+C
Tumor volume (mm3)
Days after treatment
Days
CD8 CD31 (Peri)
CD8 CD31 (Intra)
Peritumoral
Intratumoral
CD8+ area (%)
CD31+ area (%)

P, αPD-1. C, αCTLA-4. *p < 0.05 versus control; #p < 0.05 versus JX; $p < 0.05 versus αPD-1 or αCTLA-4. ns, not significant.

Figure 7. The efficacy of combination immunotherapy with intratumoral mJX-594 and ICIs is not largely affected by treatment schedule.

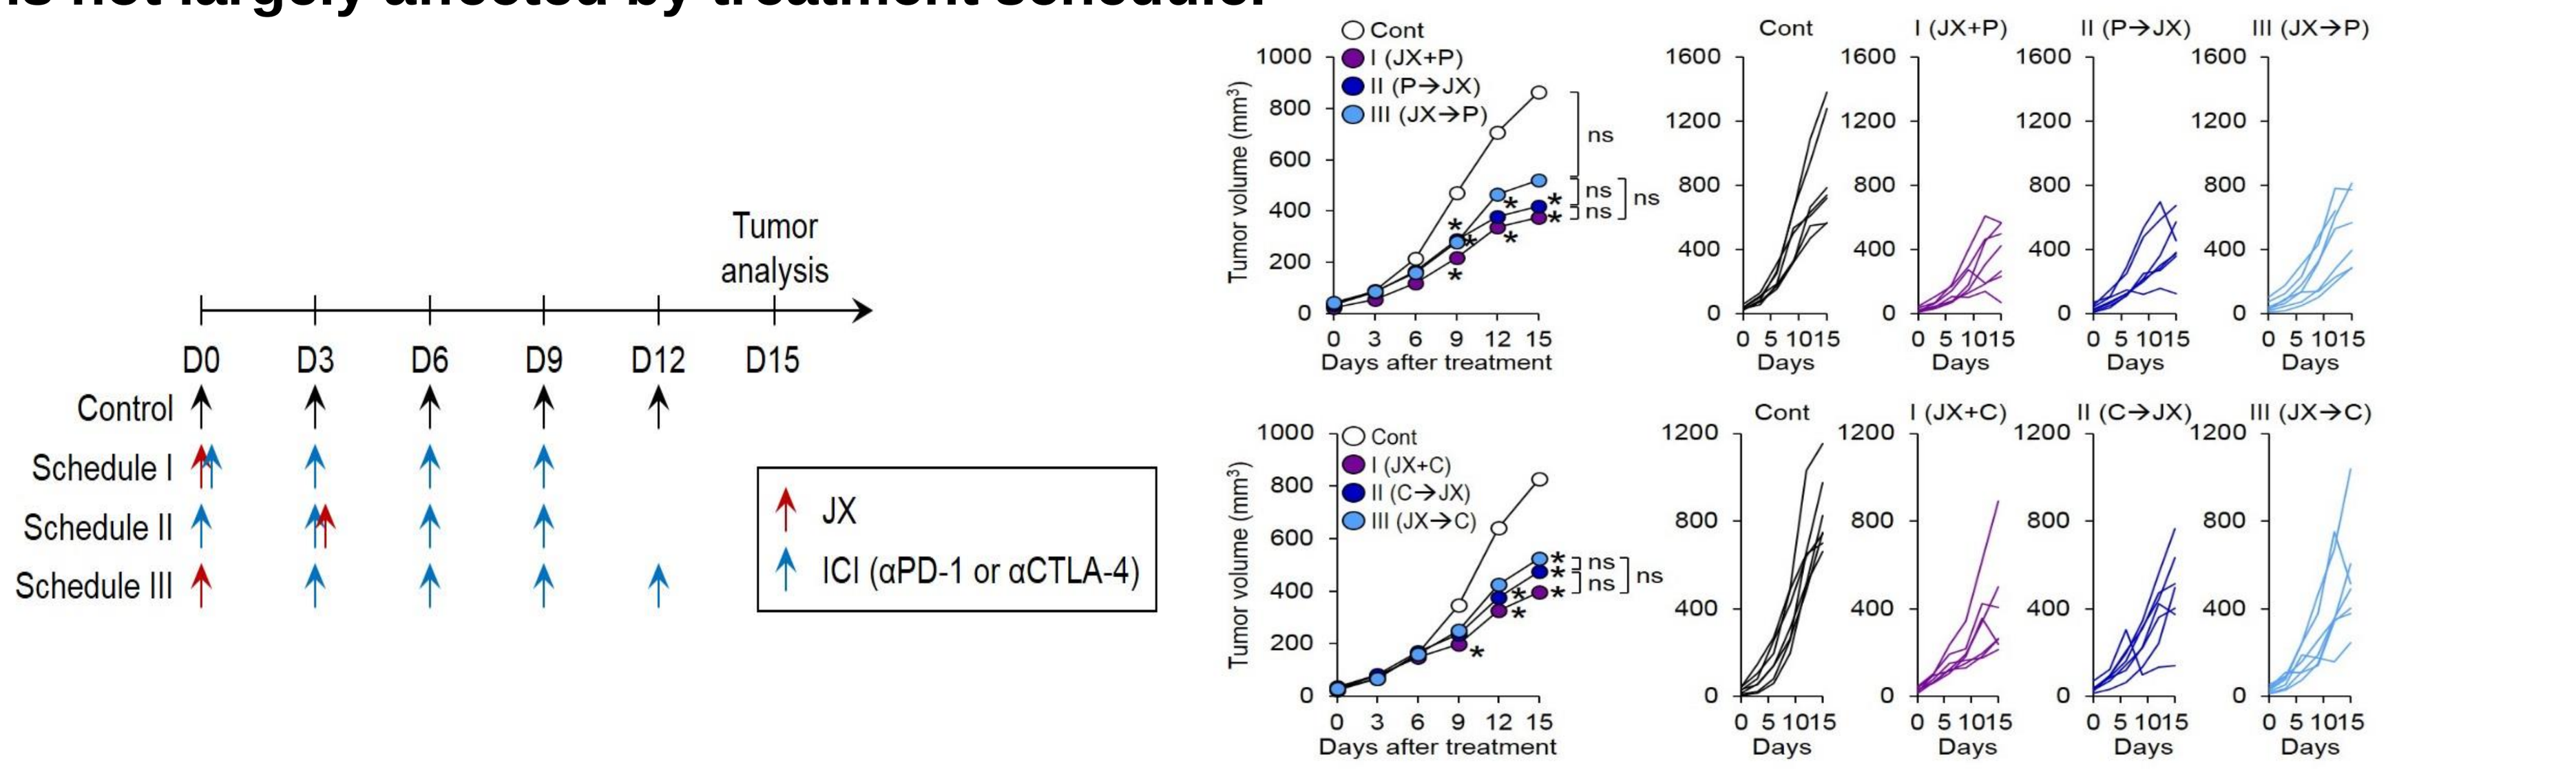
Tumor analysis
D0
D3
D6
D9
D12
D15
Control
Schedule I
Schedule II
Schedule III
JX
ICI (αPD-1 or αCTLA-4)
Cont
I (JX+P)
II (P→JX)
III (JX→P)
I (JX+C)
II (C→JX)
III (JX→C)
Tumor volume (mm3)
Days after treatment
Days

P, αPD-1. C, αCTLA-4. *p < 0.05 versus control. ns, not significant.

Figure 8. The triple combination of mJX-594, αPD1, and αCTLA4 can induce complete tumor regression and provides a long-term survival benefit in implanted kidney cancer.

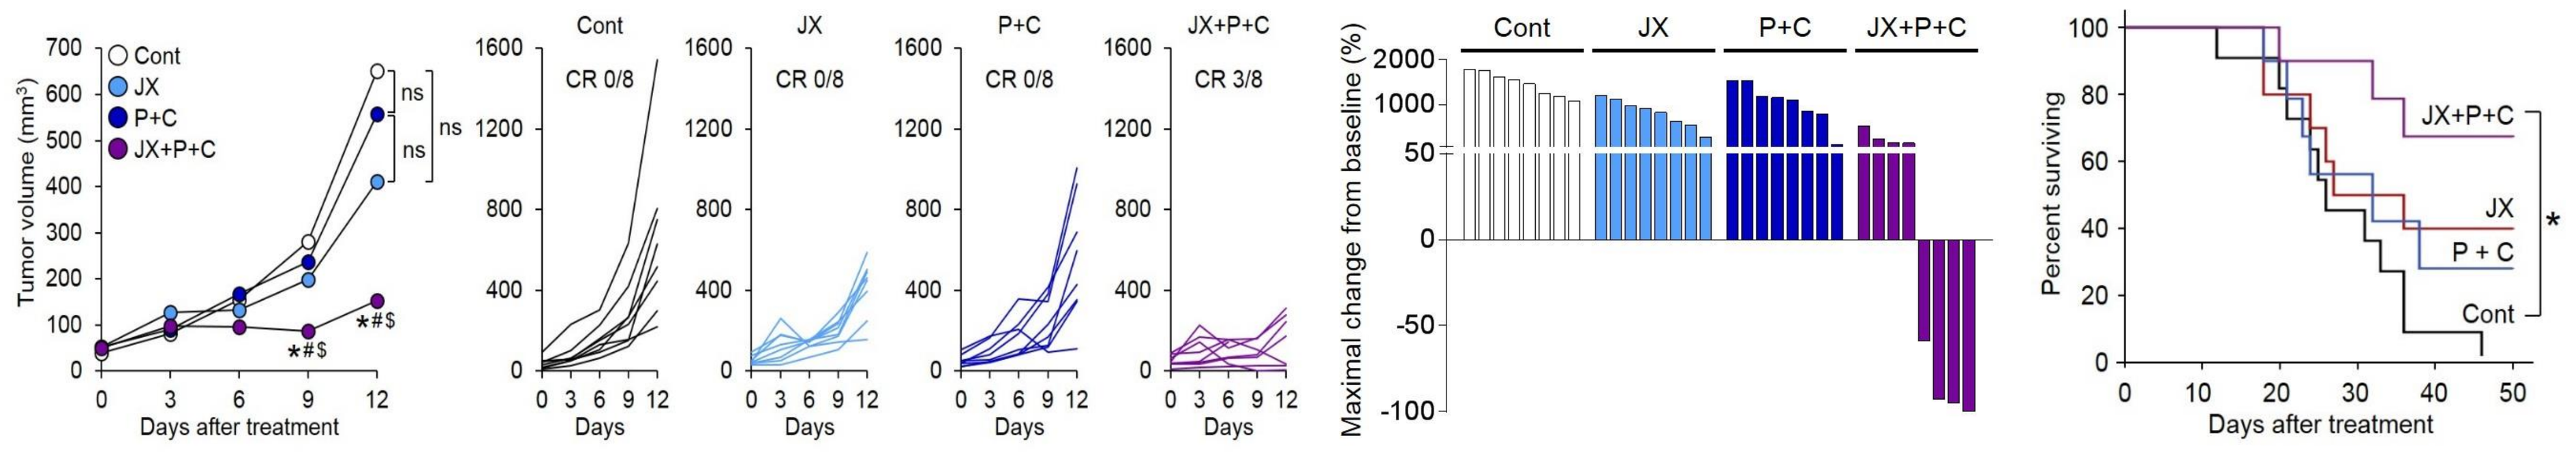
Cont
JX
P+C
JX+P+C
CR 0/8
CR 3/8
Tumor volume (mm3)
Days after treatment
Days
Maximal change from baseline (%)
Percent surviving

P, αPD-1. C, αCTLA-4. *p < 0.05 versus control; #p < 0.05 versus JX; $p < 0.05 versus αPD-1+ αCTLA-4. ns, not significant.

Introduction

In conclusion, this study indicated that intratumoral injection of mJX-954 induces a profound remodeling of TME from cold to hot state and elicit robust anti-cancer immunity in combination with ICIs, overcoming immunotherapy resistance.